Walgreens
Trusted since 1901™

Medication Synchronization Improves Medication Adherence

Presented at the International Society for Pharmacoeconomics and Outcomes Research (ISPOR); Baltimore, MD; May 19-23, 2018.

Personalized pharmacy outreach that helps patients synchronize their medications improves adherence.

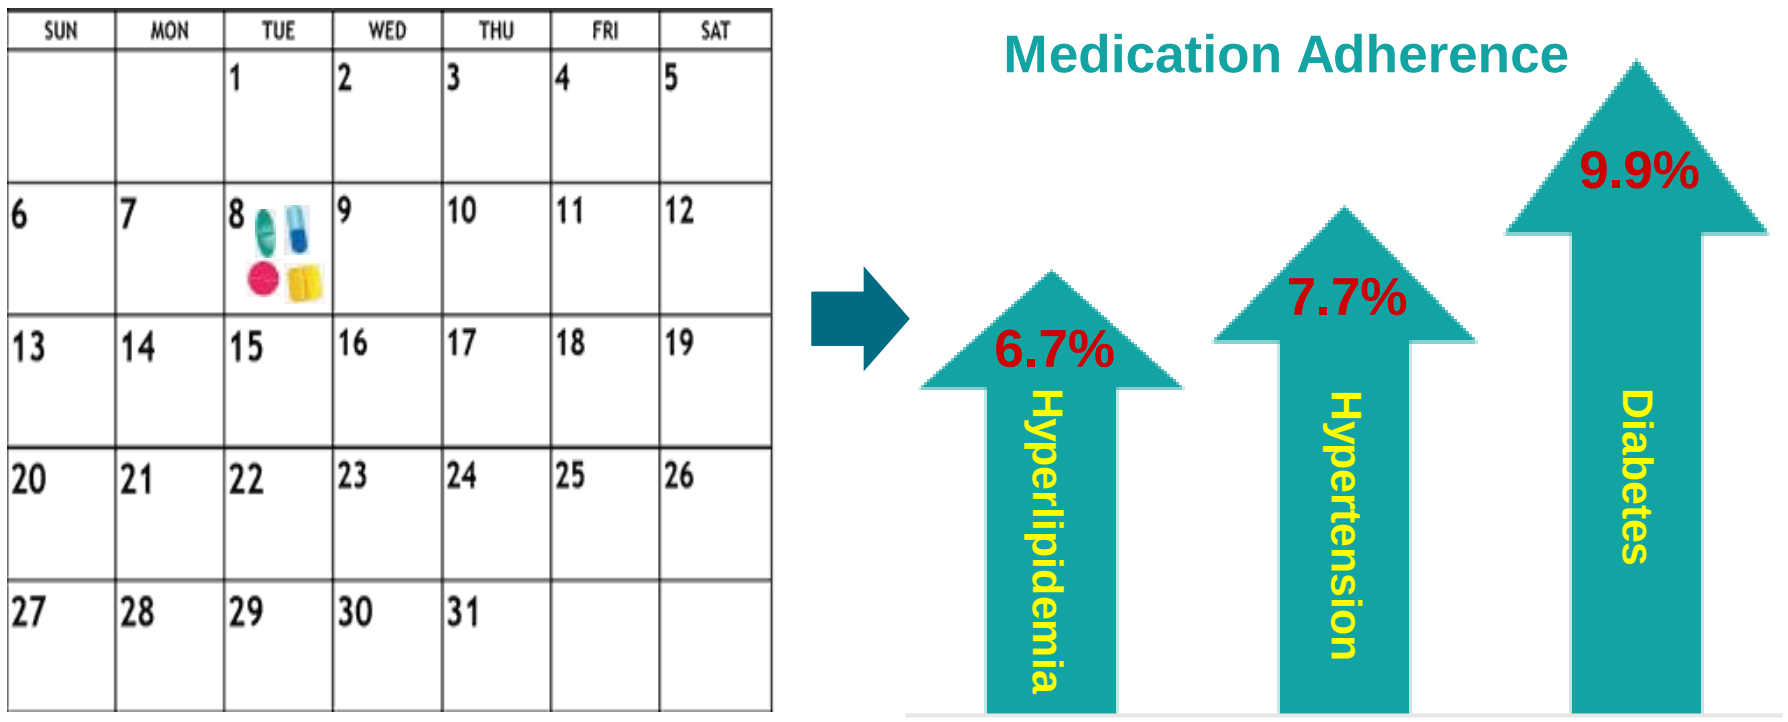
SUN MON TUE WED THU FRI SAT
Medication Adherence
6.7%
Hyperlipidemia
7.7%
Hypertension
9.9%
Diabetes

OBJECTIVE

- To evaluate the impact of a medication synchronization program on medication adherence.

METHOD

- Study Design: Retrospective study with a propensity score matched control group.
- Study Population: Patients were qualified for a medication synchronization pilot program between February and September 2016. To qualify, patients had to be 21 years and older, and in the past six months had 2 or more visits in a single month to fill 3 to 7 maintenance medications at Walgreens pharmacy. The test group included 4,221 patients who enrolled in the program at 30 pharmacies in FL, IL and NM. Patients in the control group (n=4,221) came from the same states but outside of the test stores and did not receive any synchronization service.
- Measurement Period: 12 months follow-up beginning 2-months after each patient’s qualification date to allow for initial synchronization.
- Intervention: Patients in the test group received personalized outreach calls from a central pharmacy team member for enrollment, synchronization planning and monthly confirmation. Enrolled patients receive synchronization plans which include an anchor date, a list of medications to synchronize, short fills, and quantity dispensed. Anchor date is the date agreed between patients and the pharmacy team on which all medications will be filled at the same time. Short fills which are less than the full 30 or 90 day supply may be required in order to synchronize all medications to the anchor date. The quantity dispensed (30 days or 90 days) that varies by patient and drug is displayed along with the medication. Confirmation calls are placed before the synchronization date to 1) confirm if the patient’s medications have changed; 2) realign prescriptions if necessary; 3) provide the prescription pick up date to the patient; and 4) inform the patient about future pharmacy calls.
- Outcome Measures: Medication adherence for hyperlipidemia, hypertension and diabetes were calculated and compared between test and control groups using Walgreens pharmacy claims data. It was evaluated using both proportion of days covered (PDC) and optimal adherence (PDC ≥ 80%).

©2018 Walgreen Co. All rights reserved.

- Statistical Analyses: Propensity scores were used to match patients in the control group to the test group on age, gender, plan type, copay, comorbidities, number of medications, number of pharmacy visits, and adherence at baseline. Student's t-tests and Chi-square tests were used to evaluate statistical significance of the group differences.

RESULTS

- At baseline, patients in the test group on average were 66.5 years old and 54.9% were female; they had 5.2 comorbidities and took 7.8 visits in 6 months to fill 4.4 eligible maintenance medications; average sum of copay was $101.7; total days supply was 634.9; 41.4% patients came from Florida, 36.3% from Illinois and 22.3% from New Mexico; 53.6% had Medicare Part D coverage, 26.1% commercial plan and 17.2% Medicaid; 70.9% were taking hyperlipidemia, 68.9% hypertension and 34.4% diabetes medications. The control group after propensity matching was not significantly different from the test group.
- Compared to the control group, patients in the test group had:
 - greater PDC (p<0.001) by 6.2%, 6.7%, and 8.1% for hyperlipidemia, hypertension, and diabetes medications, respectively (Figure 1);
 - higher optimal adherence (p<0.001) by 6.7%, 7.7%, and 9.9% respectively for the same drug classes (Figure 2).

Figure 1: PDC by group for hyperlipidemia, hypertension, and diabetes medications

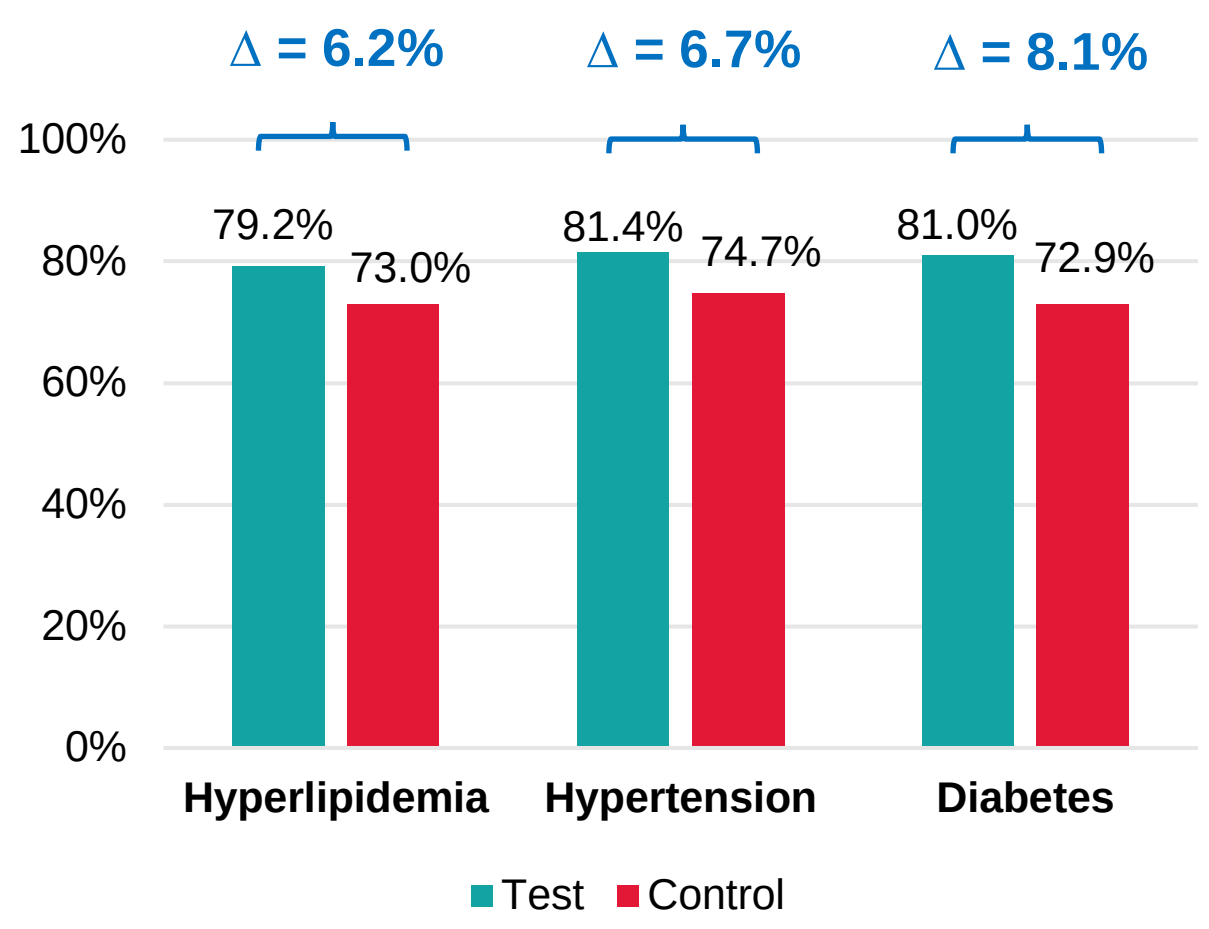

∆ = 6.2%
∆ = 6.7%
∆ = 8.1%
100%
80%
60%
40%
20%
0%
79.2%
73.0%
81.4%
74.7%
81.0%
72.9%
Hyperlipidemia
Hypertension
Diabetes
Test
Control

Figure 2: Percent of optimally adherent patients by group for hyperlipidemia, hypertension, and diabetes medications

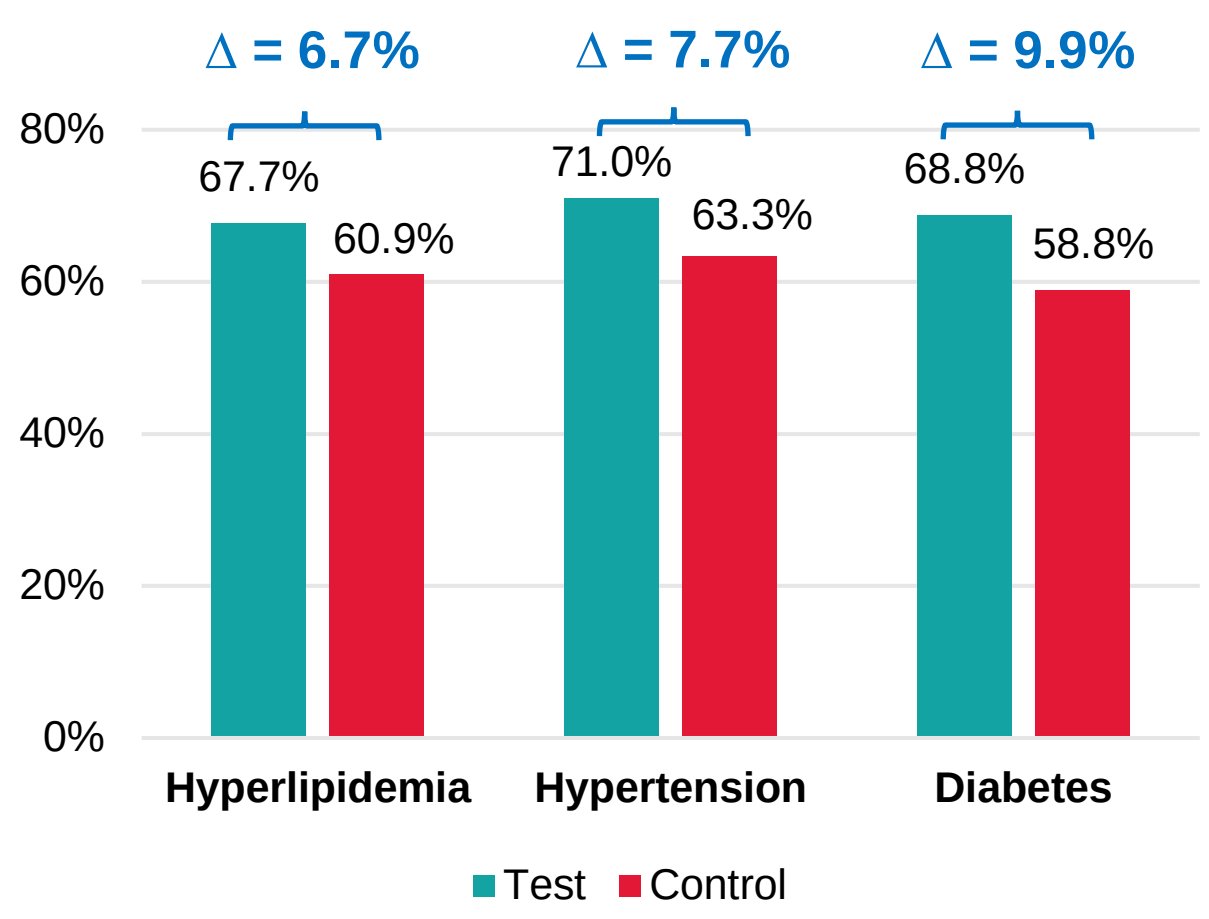

∆ = 6.7%
∆ = 7.7%
∆ = 9.9%
80%
60%
40%
20%
0%
67.7%
60.9%
71.0%
63.3%
68.8%
58.8%
Hyperlipidemia
Hypertension
Diabetes
Test
Control

CONCLUSIONS

- Medication synchronization helps patients with multiple chronic conditions improve their medication adherence, which could result in better health outcomes and reduced overall healthcare cost.

AMA Citation:
Mu, Y, Lou, Y, Taitel, M. Medication synchronization improves medication adherence. Presented at the International Society for Pharmacoeconomics and Outcomes Research conference, May 19-23, 2018, Baltimore, MD.

Contributing Authors:
Ying Mu, MS[1]; Youbei Lou, PhD[1]; Michael Taitel, PhD[1]
1 Walgreens Center for Health and Wellbeing Research

This research was approved by Quorum IRB (#32557). This research was funded internally by Walgreen Co.

For more information on this presentation, please contact: research@walgreens.com.

©2018 Walgreen Co. All rights reserved.
2